A Randomized, Double-Blind, Parallel-Group, Placebo-Controlled Phase III Trial to Evaluate Efficacy and Safety of Ocrelizumab in Primary Progressive Multiple Sclerosis

X Montalban,[1] B Hemmer,[2,3] K Rammohan,[4] G Giovannoni,[5] J de Seze,[6] A Bar-Or,[7] DL Arnold,[7,8] A Sauter,[9] D Masterman,[10] P Fontoura,[9] P Chin,[10] H Garren,[9] J Wolinsky,[11]
on behalf of the ORATORIO clinical investigators

[1]Hospital Vall d'Hebron University, Barcelona, Spain; [2]Technische Universität München, Munich, Germany; [3]Munich Cluster for Systems Neurology (SyNergy), Munich, Germany; [4]University of Miami, Miami, FL, USA; [5]Queen Mary University of London, London, UK; [6]University Hospital of Strasbourg, Strasbourg, France; [7]McGill University, Montreal, QC, Canada; [8]NeuroRx Research, Montreal, QC, Canada; [9]F. Hoffmann-La Roche Ltd., Basel, Switzerland; [10]Genentech, Inc., San Francisco, CA, USA; [11]University of Texas Health Science Center at Houston, Houston, TX, USA

BACKGROUND

- Patients with primary progressive multiple sclerosis (PPMS) experience disability progression from disease onset[1]
- Previous PPMS trials have failed to demonstrate significant treatment effect in slowing the course of disability progression[2]
- A better understanding of the pathophysiology of PPMS disease progression is required, and an approved treatment with a favorable benefit/risk balance is an important unmet need[2-4]
- Ocrelizumab (OCR) is a humanized monoclonal antibody that selectively depletes CD20+ B cells, while preserving the capacity for B-cell reconstitution and preexisting humoral immunity

METHODS

Study design

- Patients were randomized (2:1) to receive OCR 600 mg, given as two 300 mg intravenous infusions 14 days apart, or matching placebo every 24 weeks for ≥120 weeks until an overall prespecified number of confirmed disability progression (CDP) events occurred (**Figure 1**)
 - — The double-blind treatment period was designed to end when approximately 253 events were reached, based on the original sample size assumptions
 - — If the number of events had not been reached by 120 weeks after the last patient was randomized, the study would continue until the target number of events had been reached
- Eligible patients were stratified by age (≤45 vs >45 years) and region (US vs rest of world)
- Patients discontinuing treatment entered safety follow-up ≥48 weeks from the date of the last infusion and underwent B cell monitoring for repletion

Figure 1: ORATORIO study design

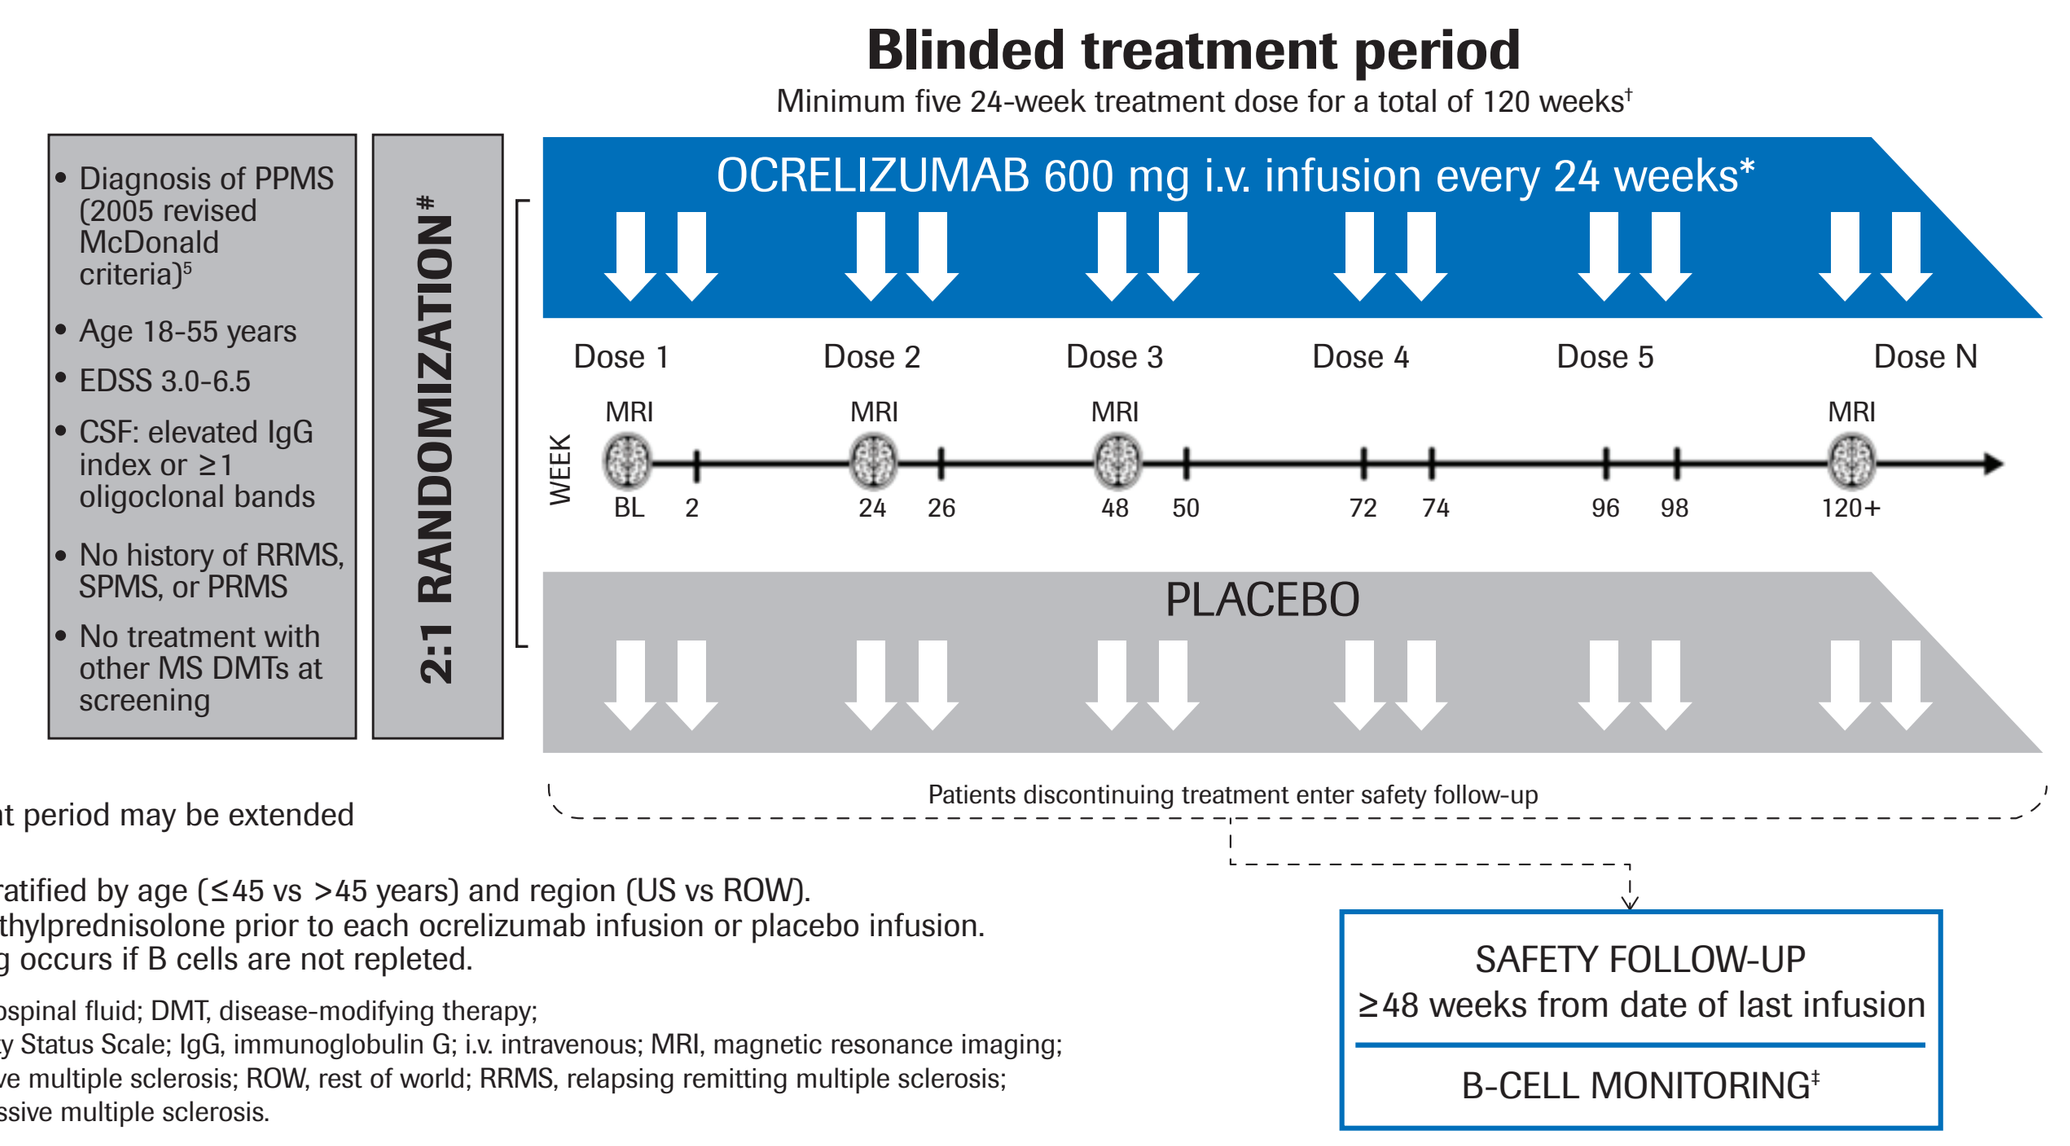
Blinded treatment period
Minimum five 24-week treatment dose for a total of 120 weeks†
• Diagnosis of PPMS (2005 revised McDonald criteria)[5]
• Age 18-55 years
• EDSS 3.0-6.5
• CSF: elevated IgG index or ≥1 oligoclonal bands
• No history of RRMS, SPMS, or PRMS
• No treatment with other MS DMTs at screening
2:1 RANDOMIZATION#
OCRELIZUMAB 600 mg i.v. infusion every 24 weeks*
Dose 1
Dose 2
Dose 3
Dose 4
Dose 5
Dose N
MRI
WEEK
BL 2 24 26 48 50 72 74 96 98 120+
PLACEBO
Patients discontinuing treatment enter safety follow-up
SAFETY FOLLOW-UP
≥48 weeks from date of last infusion
B-CELL MONITORING‡

†The blinded treatment period may be extended until database lock.
#2:1 randomization stratified by age (≤45 vs >45 years) and region (US vs ROW).
*Patients received methylprednisolone prior to each ocrelizumab infusion or placebo infusion.
‡Continued monitoring occurs if B cells are not repleted.

BL, baseline; CSF, cerebrospinal fluid; DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; IgG, immunoglobulin G; i.v. intravenous; MRI, magnetic resonance imaging; PPMS, primary progressive multiple sclerosis; ROW, rest of world; RRMS, relapsing remitting multiple sclerosis; SPMS, secondary progressive multiple sclerosis.

Study endpoints

- Primary endpoint
 - — Time to onset of CDP sustained for ≥12 weeks
- Key secondary endpoints
 - — Time to onset of CDP sustained for ≥24 weeks
 - — Change in timed 25-foot walk (T25FW; baseline to Week 120)
 - — Change in total volume of brain T2 hyperintense lesions (baseline to Week 120)
 - — Change in whole brain volume (Week 24 to Week 120)
 - — Safety and tolerability of OCR 600 mg intravenously every 24 weeks in patients with PPMS

Statistical analysis

- All efficacy analyses were performed on the intent-to-treat (ITT) population
- For time to onset of 12- and 24-week CDP, the p-values were based on a log-rank test stratified by geographic region and age. The overall hazard ratio was estimated using a stratified Cox regression model with the same stratification factors used in the log-rank test. Patients with initial disability progression who discontinued treatment early with no confirmatory EDSS assessment were considered as having confirmed disability
- For T25FW and total volume of brain T2 hyperintense lesions, the p-values were based on ranked ANCOVA at the 120-week visit adjusted for baseline, geographic region and age, with missing values imputed by last observation carried forward. Point estimates and 95% confidence intervals were calculated using a mixed-effect model repeated measure (MMRM) analysis on log-transformed data adjusted for baseline, geographic region and age
- For change in whole brain volume, the p-value is based on MMRM on percentage change adjusted for week 24 brain volume, geographic region and age
- Secondary efficacy endpoints were tested in hierarchical order at a two-sided alpha of 0.05; from the first p-value above 0.05, all p-values following in the predetermined hierarchy became nonconfirmatory

RESULTS

Baseline demographics and disease characteristics

- The treatment arms were well balanced (**Table 1**)

Table 1: Baseline demographics and disease characteristics

	Placebo (n=244)	Ocrelizumab (n=488)
Age, yrs, mean (SD)	44.4 (8.3)	44.7 (7.9)
Female, n (%)	124 (50.8)	237 (48.6)
Time since MS symptom onset, yrs, mean (SD)	6.1 (3.6)	6.7 (4.0)
Time since MS diagnosis, yrs, mean (SD)	2.8 (3.3)	2.9 (3.2)
MS disease-modifying treatment naive,* n (%)	214 (87.7)	433 (88.7)
EDSS, mean (SD)	4.7 (1.2)	4.7 (1.2)
MRI		
Patients with Gd+ lesions, n (%)	60 (24.7)	133 (27.5)
Number of Gd+ T1 lesions, mean (SD)	0.6 (1.6)	1.2 (5.1)
T2 lesion volume, cm³, mean (SD)	10.9 (13.0)	12.7 (15.1)
Normalized brain volume, cm³, mean (SD)	1469.9 (88.7)	1462.9 (83.9)

*No disease-modifying treatments in the previous 2 years;
EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; MRI, magnetic resonance imaging, MS, multiple sclerosis; SD, standard deviation.

Study completion

- The mean treatment duration was approximately 3 years
- 390 of treated patients (80%) in the OCR arm remained on treatment at the clinical cut-off date compared with 159 (67%) in the placebo arm
- Of patients withdrawn from the study treatment at the clinical cut-off date, 61 (64%) in the OCR arm entered safety follow-up, compared with 45 (56%) in the placebo arm

Disability progression

- Compared with placebo, OCR significantly reduced the risk of 12-week CDP by 24% (p=0.0321; **Figure 2A**) and the risk of 24-week CDP by 25% (p=0.0365; **Figure 2B**)
- OCR reduced the progression rate of ambulation impairment as measured by change in T25FW at Week 120 by 29% (p=0.0404) compared with placebo (**Figure 3**)

Figure 2: Time to onset of disability progression confirmed after ≥12 weeks (A) and ≥24 weeks (B)

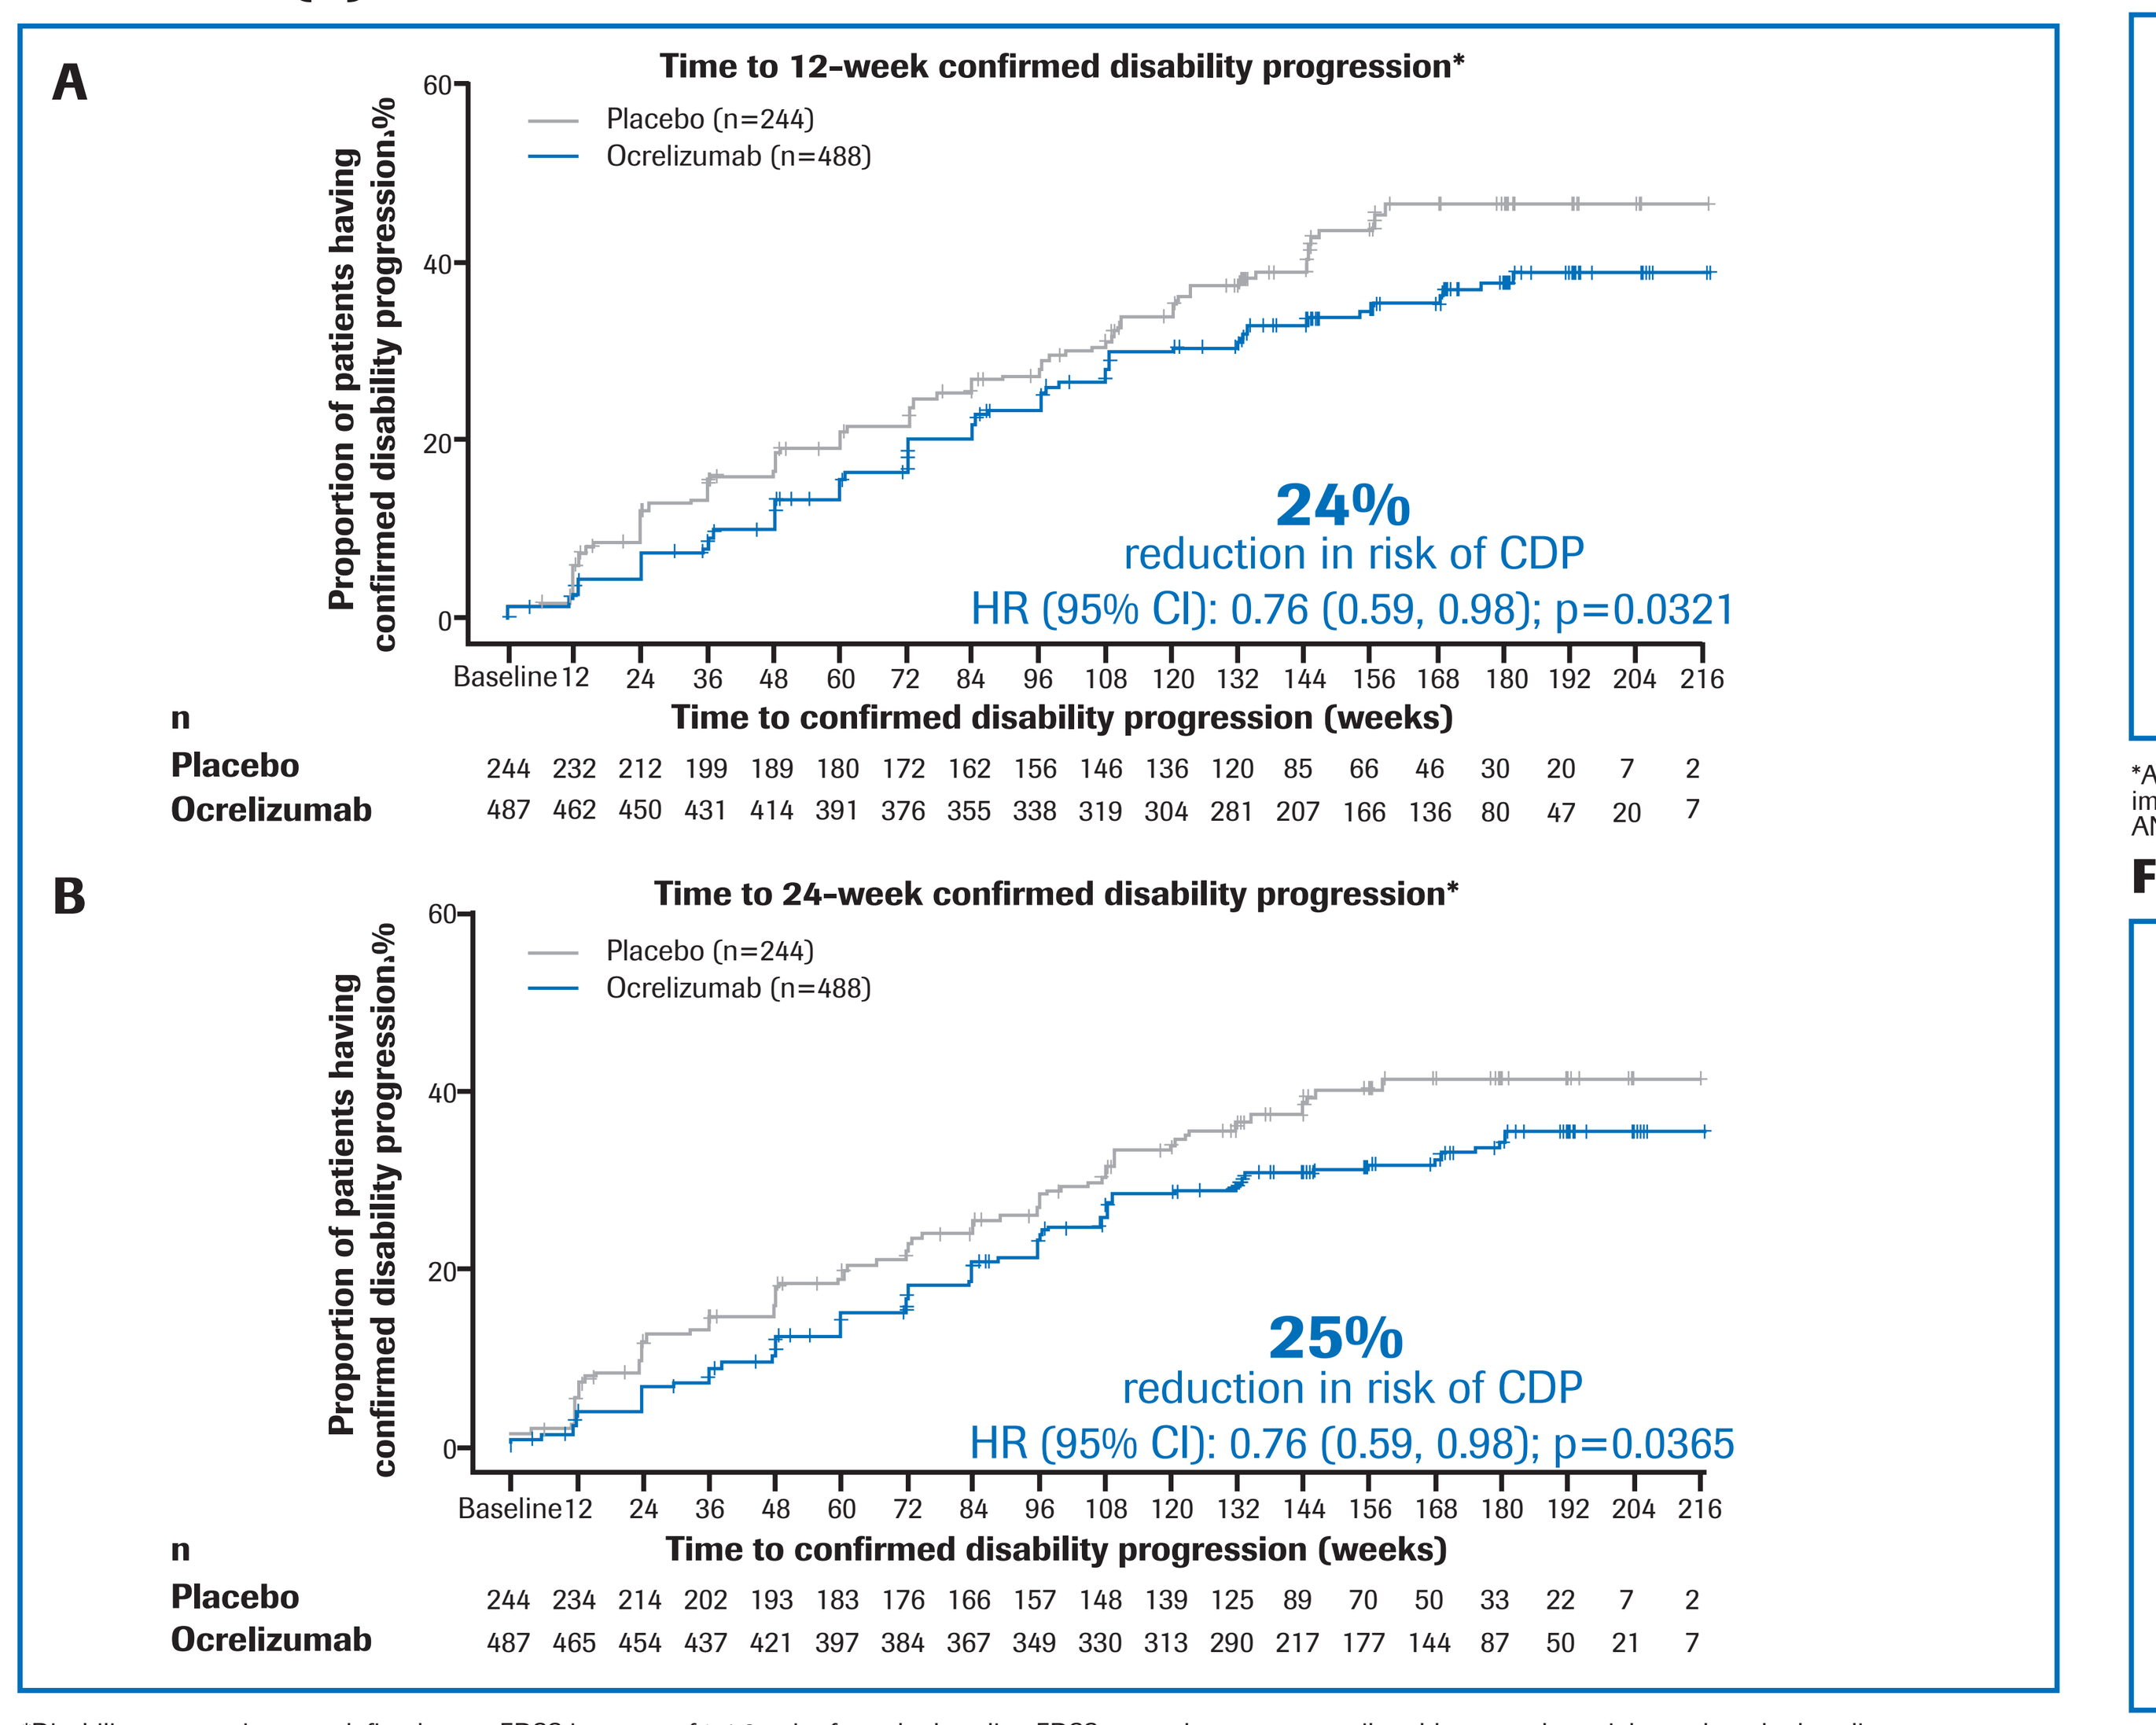
A
Time to 12-week confirmed disability progression*
Placebo (n=244)
Ocrelizumab (n=488)
Proportion of patients having confirmed disability progression,%
60
40
20
0
24%
reduction in risk of CDP
HR (95% CI): 0.76 (0.59, 0.98); p=0.0321
Baseline 12 24 36 48 60 72 84 96 108 120 132 144 156 168 180 192 204 216
Time to confirmed disability progression (weeks)
n
Placebo 244 232 212 199 189 180 172 162 156 146 136 120 85 66 46 30 20 7 2
Ocrelizumab 487 462 450 431 414 391 376 355 338 319 304 281 207 166 136 80 47 20 7
B
Time to 24-week confirmed disability progression*
Placebo (n=244)
Ocrelizumab (n=488)
Proportion of patients having confirmed disability progression,%
60
40
20
0
25%
reduction in risk of CDP
HR (95% CI): 0.76 (0.59, 0.98); p=0.0365
Baseline 12 24 36 48 60 72 84 96 108 120 132 144 156 168 180 192 204 216
Time to confirmed disability progression (weeks)
n
Placebo 244 234 214 202 193 183 176 166 157 148 139 125 89 70 50 33 22 7 2
Ocrelizumab 487 465 454 437 421 397 384 367 349 330 313 290 217 177 144 87 50 21 7

*Disability progression was defined as an EDSS increase of ≥1.0 point from the baseline EDSS score that was not attributable to another etiology when the baseline score was ≤5.5, and ≥0.5 point when the baseline score was >5.5. Analysis based on ITT population; p-value based on log-rank test stratified by geographic region and age. Patients with initial disability progression who discontinued treatment early with no confirmatory EDSS assessment were considered as having confirmed disability progression.
CDP, confirmed disability progression; CI, confidence interval; EDSS, Expanded Disability Status Scale; HR, hazard ratio; ITT, intent to treat.

Figure 3: Change in timed 25-foot walk from baseline to Week 120

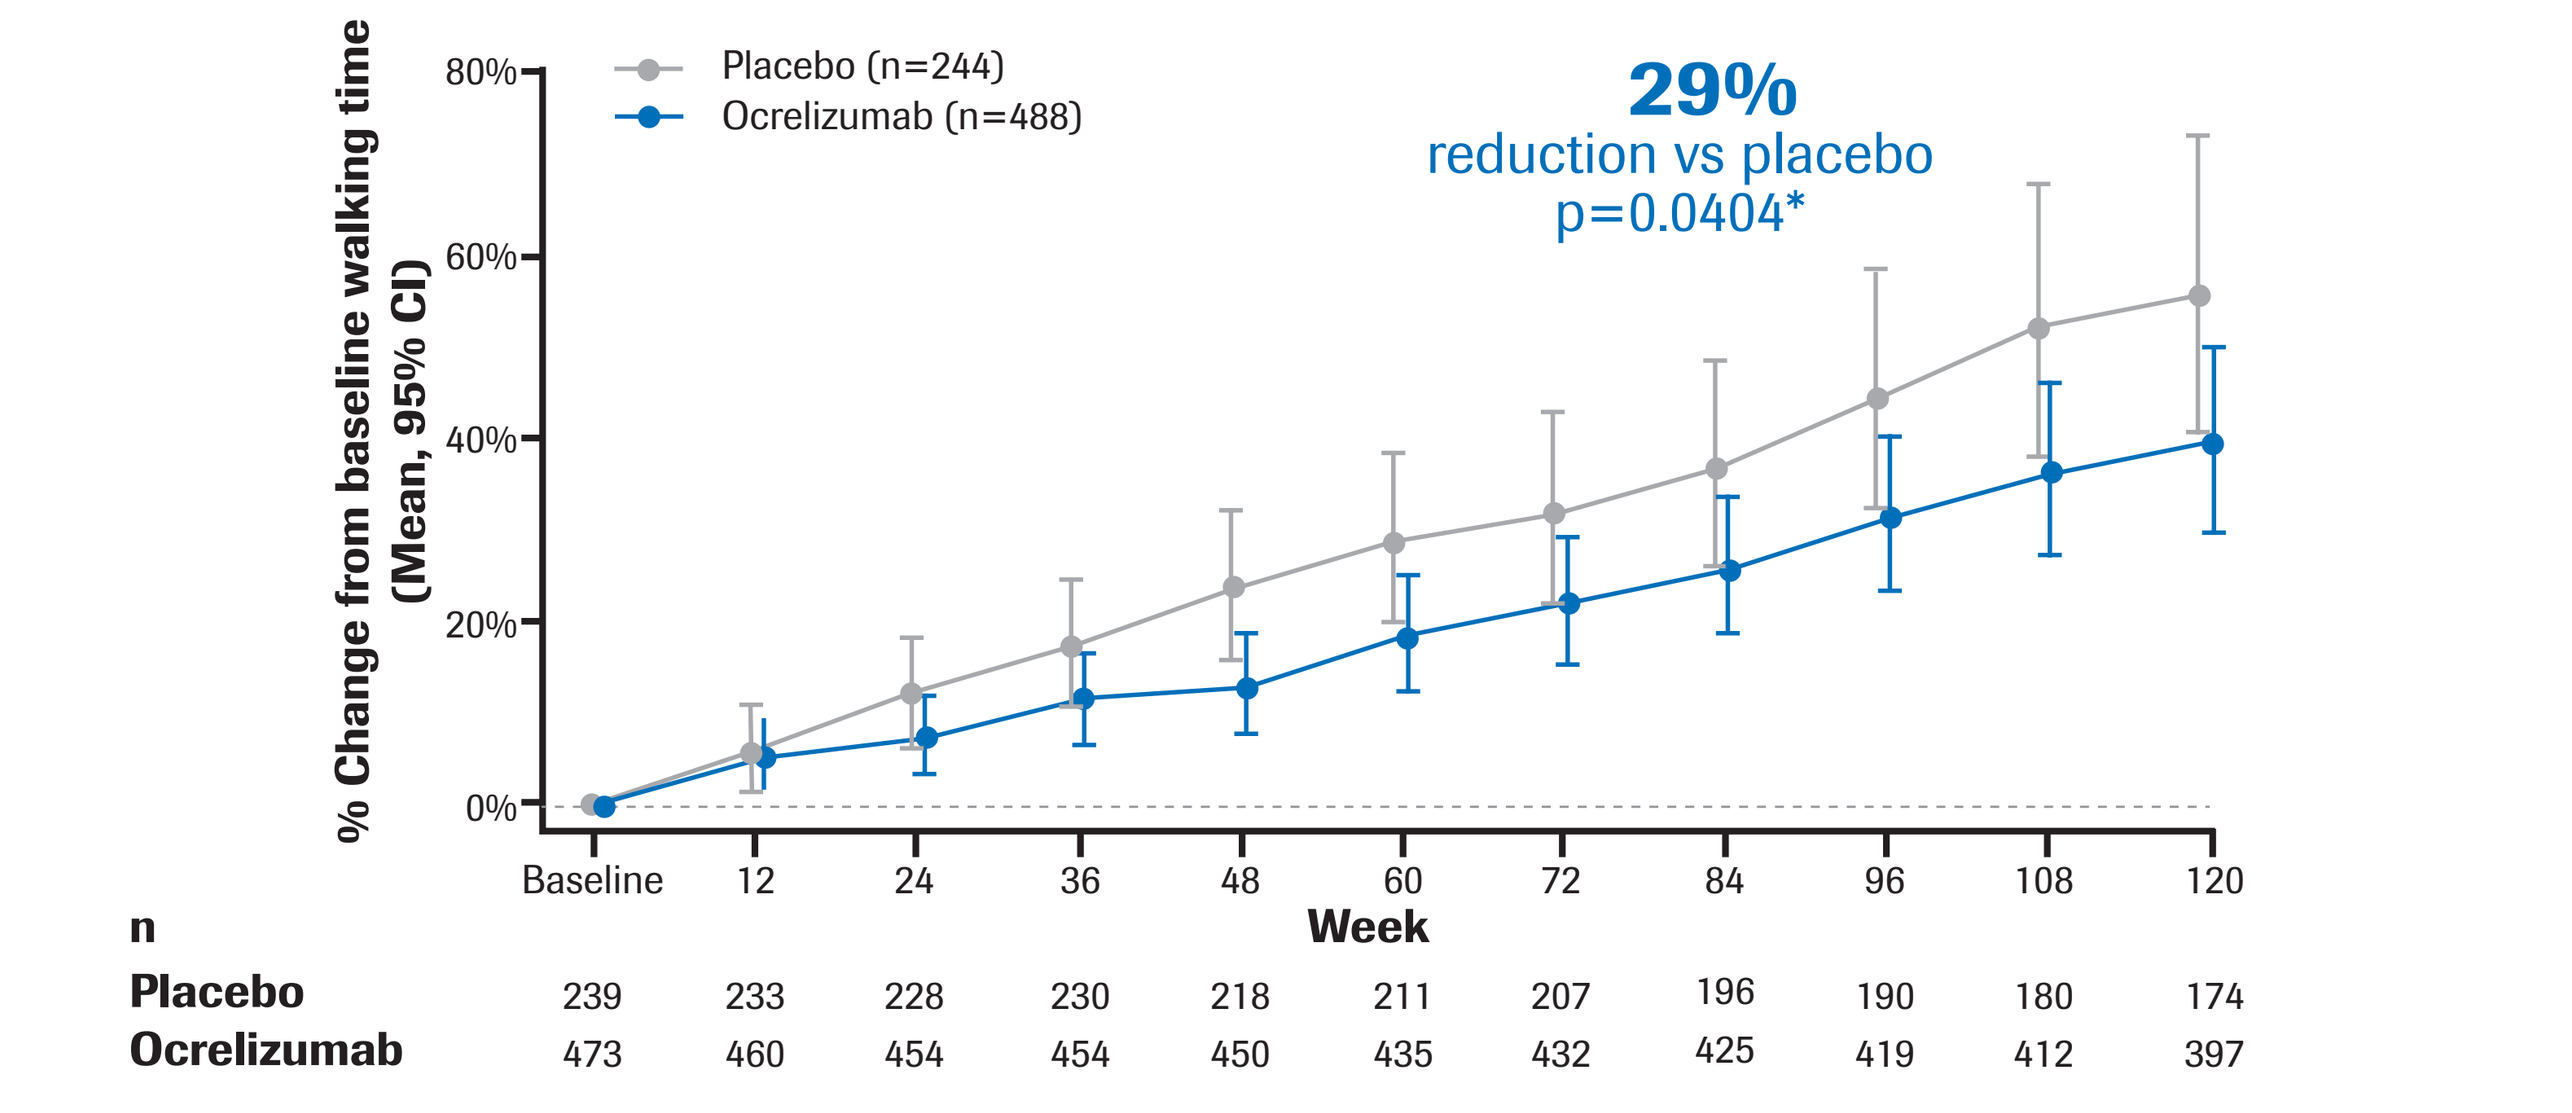
Placebo (n=244)
Ocrelizumab (n=488)
29%
reduction vs placebo
p=0.0404*
% Change from baseline walking time (Mean, 95% CI)
80%
60%
40%
20%
0%
Baseline 12 24 36 48 60 72 84 96 108 120
Week
n
Placebo 239 233 228 230 218 211 207 196 190 180 174
Ocrelizumab 473 460 454 454 450 435 432 425 419 412 397

*Analysis based on ITT population; p-value based on ranked ANCOVA at 120-week visit adjusted for baseline timed 25-foot walk, geographic region and age, with missing values imputed by LOCF.
Point estimates and 95% CI based on mixed-effect model repeated measures analysis on log-transformed data adjusted for baseline timed 25-foot walk, geographic region, and age.
ANCOVA, analysis of covariance; CI, confidence intervals; ITT, intent to treat; LOCF, last observation carried forward; MMRM, mixed-effect model repeated measures.

Brain MRI endpoints

- OCR reduced the total volume of brain T2 hyperintense lesions from baseline to Week 120, whereas the total lesion volume increased at Week 120 compared with baseline in the placebo group (p<0.0001; **Figure 4**)
 - — In the placebo group, brain T2 hyperintense lesion volume increased by 7.4%
 - — OCR decreased brain T2 hyperintense lesion volume by 3.4%
- Compared with placebo, OCR reduced the rate of whole brain volume loss from Week 24 to Week 120 by 17.5% (p=0.0206; **Figure 5**)

Figure 4: Change in brain T2 hyperintense lesion volume from baseline to Week 120

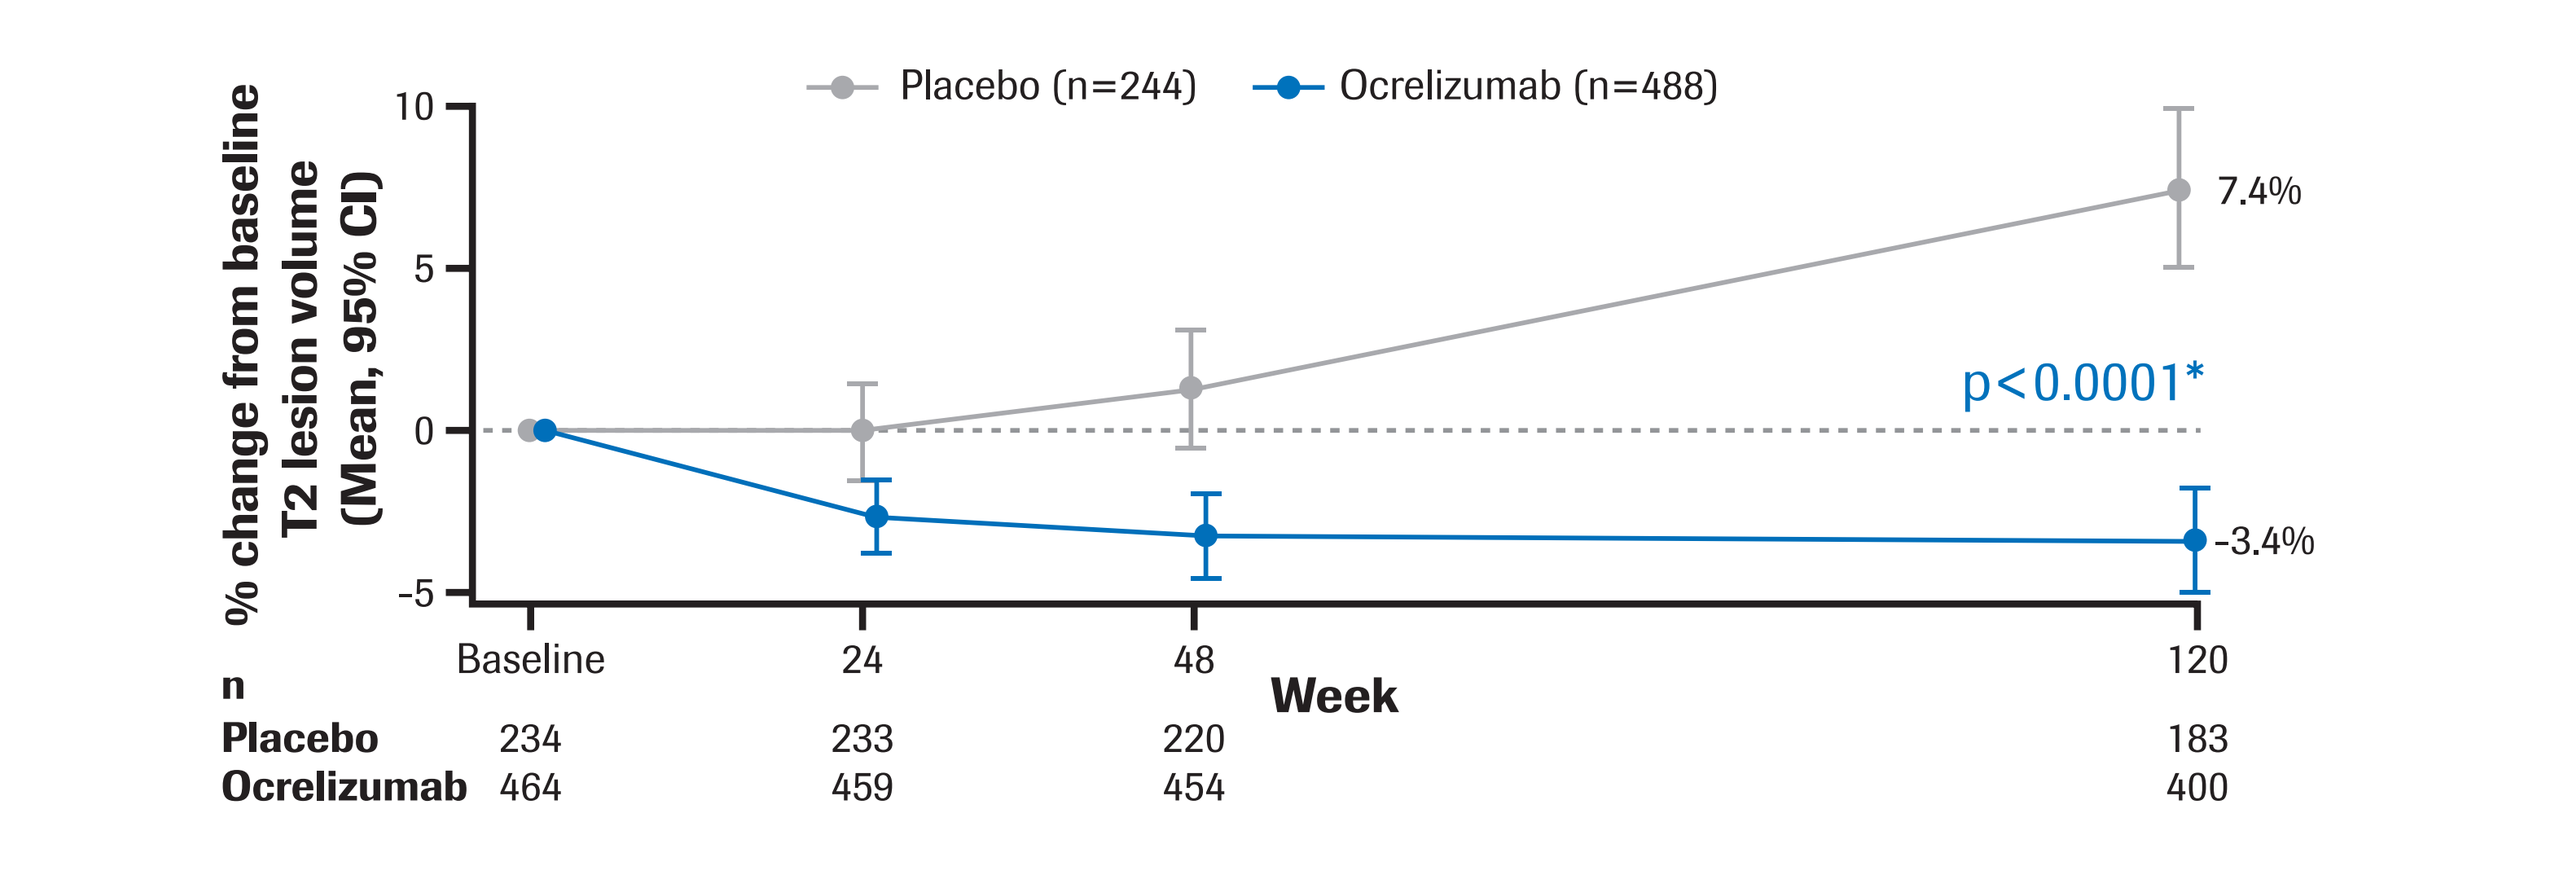
Placebo (n=244)
Ocrelizumab (n=488)
% change from baseline T2 lesion volume (Mean, 95% CI)
10
5
0
−5
7.4%
p<0.0001*
-3.4%
Baseline 24 48 120
Week
n
Placebo 234 233 220 183
Ocrelizumab 464 459 454 400

*Analysis based on ITT population; p-value based on ranked ANCOVA at 120-week visit adjusted for baseline T2 lesion volume, geographic region, and age, with missing values imputed by LOCF. Point estimates and 95% CI based on MMRM analysis on log-transformed data adjusted for baseline T2 lesion volume, geographic region, and age.
ANCOVA, analysis of covariance; CI, confidence interval; ITT, intent to treat; LOCF, last observation carried forward; MMRM, mixed-effect model repeated measures.

Figure 5: Change in whole brain volume from Week 24 to Week 120

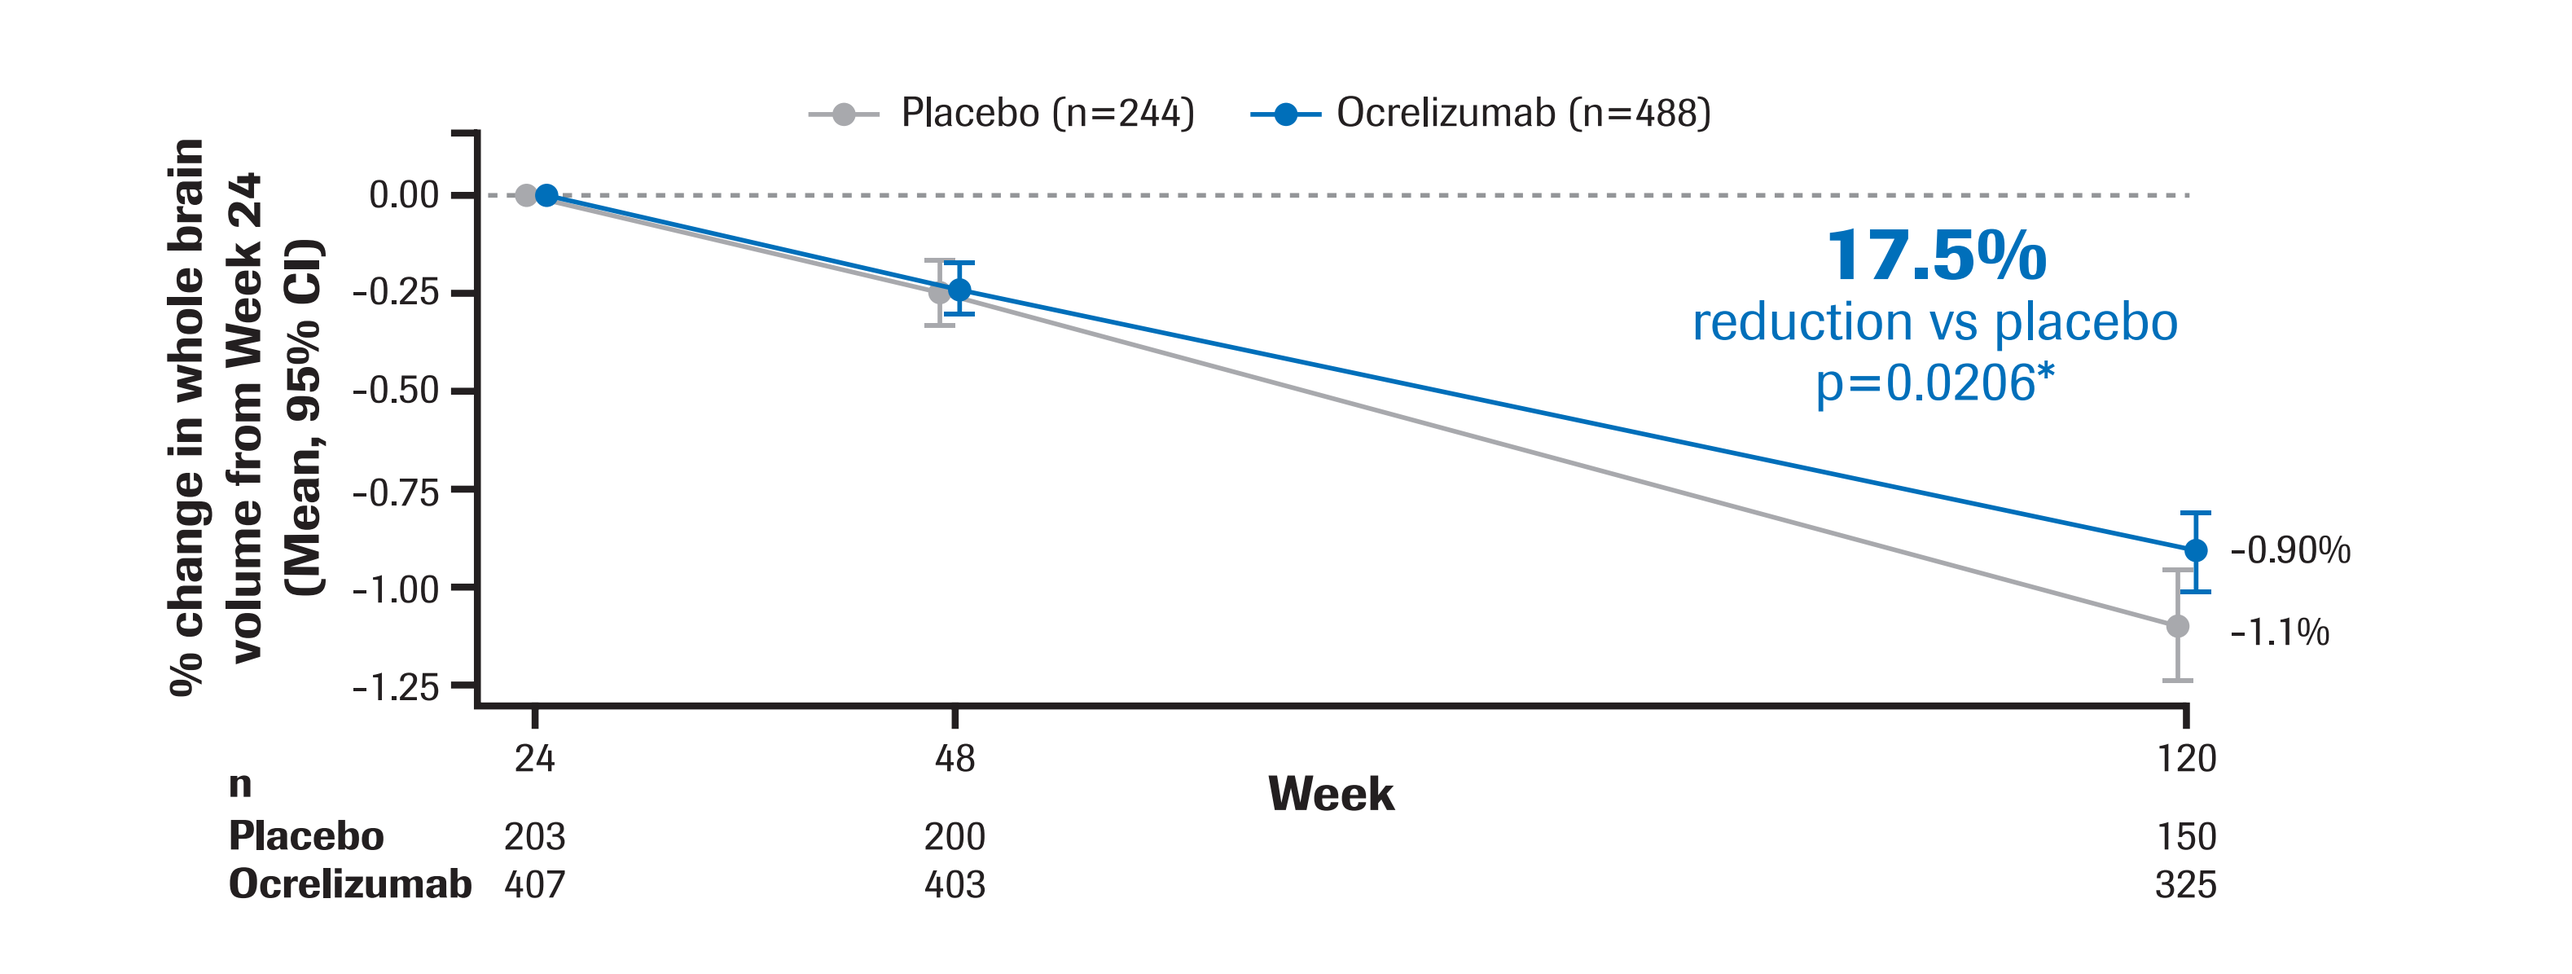
Placebo (n=244)
Ocrelizumab (n=488)
% change in whole brain volume from Week 24 (Mean, 95% CI)
0.00
-0.25
-0.50
-0.75
-1.00
-1.25
17.5%
reduction vs placebo
p=0.0206*
-0.90%
-1.1%
24 48 120
Week
n
Placebo 203 200 150
Ocrelizumab 407 403 325

*Analysis based on ITT population with Week 24 and at least one post-Week 24 assessment; p-value based on MMRM at 120-week visit adjusted for Week 24 brain volume, geographic region and age.
CI, confidence interval; ITT, intent to treat; MMRM, mixed-effect model repeated measures.

Safety

- The proportion of patients reporting adverse events (AEs) was 95.1% and 90.0% in the OCR and placebo groups, respectively (**Table 2**)
 - — A higher proportion of patients treated with OCR reported upper respiratory tract infections (10.9%) compared with placebo (5.9%)
 - — Oral herpes infection was more common among patients treated with OCR (2.3%) compared with placebo (0.4%)
- Infusion-related reactions (IRRs) included all events occurring during infusion, shortly post-infusion (in clinic) or within 24 hours post-infusion
 - — IRRs were the most frequently reported AE among OCR-treated patients; overall, 39.9% of OCR-treated patients and 25.5% patients receiving placebo reported at least one IRR
 - — A higher proportion of patients experienced IRRs of any grade with OCR (27.4%) compared with placebo (12.1%) during the first infusion of the first dose
- No fatal or life-threatening IRRs have been reported, and most IRRs were of mild to moderate severity, decreasing in both rate and severity with subsequent dosing
 - — All IRRs were manageable through premedication, infusion adjustment, and symptomatic treatment
 - — 0.4% withdrew from OCR treatment due to IRRs; 1 patient experienced an IRR at the first infusion of the first dose and the other patient at the first infusion of the second dose
- Serious AEs were reported in 20.4% of OCR-treated patients and 22.2% of patients receiving placebo (**Table 2**)
- Thirteen patients reported malignancy:
 - — 0.8% in the placebo arm: 1 reported a cervix adenocarcinoma *in situ* and 1 reported a basal cell carcinoma
 - — 2.3% in the OCR arm: 4 female patients reported a breast cancer (invasive ductal breast carcinoma), 1 reported an endometrial adenocarcinoma, 1 reported an anaplastic large-cell lymphoma (mainly T cells), 1 reported a malignant fibrous histiocytoma, 1 reported a metastatic pancreas cancer, and 3 patients reported a basal cell carcinoma (1 patient reported 3 lesions)
- Five deaths were reported:
 - — 0.4% in the placebo arm: road traffic accident
 - — 0.8% in the OCR arm: pulmonary embolism, pneumonia, pancreas carcinoma, and pneumonia aspiration

Table 2: Summary of safety in overall study population

n (%)	Placebo (n=239)	Ocrelizumab (n=486)
Overall patients with ≥1 AE	215 (90.0)	462 (95.1)
Patients with infections and infestations	162 (67.8)	339 (69.8)
Overall patients with ≥1 SAE	53 (22.2)	99 (20.4)
Patients with ≥1 serious infection event	14 (5.9)	30 (6.2)
Pneumonia	2 (0.8)	6 (1.2)
Urinary tract infection	2 (0.8)	5 (1.0)
Urosepsis	3 (1.3)	2 (0.4)
Appendicitis	0	2 (0.4)
Bronchitis	0	2 (0.4)
Colitis	0	2 (0.4)
Infectious colitis	1 (0.4)	1 (0.2)
Pyelonephritis	0	2 (0.4)
Abscess of limb	0	1 (0.2)
Abscess of eyelid	1 (0.4)	0
Appendicitis perforated	1 (0.4)	0
Arthritis infective	1 (0.4)	0
Bacterial pyelonephritis	0	1 (0.2)
Bronchopneumonia	0	1 (0.2)
Bursitis infective	0	1 (0.2)
Diverticulitis	0	1 (0.2)
Erysipelas	0	1 (0.2)
Total number of deaths	1 (0.4)	4 (0.8)
Overall patients with malignancies	2 (0.8)	11 (2.3)

AE, adverse event; SAE, serious adverse event.

CONCLUSIONS

- **In ORATORIO, when compared with placebo, OCR consistently improved both clinical and brain imaging measures of disease progression**
- **Complete safety analyses are ongoing, including investigation of numerical imbalance in malignancies; however, due to the low number of malignancies, differences must be considered within the context of the full OCR Phase III program data, including OPERA I and OPERA II trials in relapsing MS[6]**
- **OCR is the first investigational treatment to meet primary and key secondary efficacy endpoints in a Phase III PPMS study**

DISCLOSURES

X Montalban has received speaking honoraria and travel expense reimbursement for participation in scientific meetings, has been a steering committee member of clinical trials, or participated in advisory boards of clinical trials in the past years with Actelion, Almirall, Bayer, Biogen, Genzyme, Merck, Novartis, Octapharma, Receptos, F. Hoffmann-La Roche Ltd., Sanofi, Teva, and Trophos; B Hemmer has served on scientific advisory boards for F. Hoffmann-La Roche Ltd., Novartis, Bayer Schering, Merck Serono, Biogen, GSK, Chugai Pharmaceuticals Co., Ltd., Micromet, Genentech, Inc., and Genzyme Corporation; serves on the international advisory board of Archives of Neurology, Multiple Sclerosis Journal, and Experimental Neurology; has received speaker honoraria from Bayer Schering, Novartis, Biogen, Merck Serono, F. Hoffmann-La Roche Ltd., and Teva Pharmaceutical Industries Ltd.; and has received research support from Biogen, Bayer Schering, Merck Serono, Five Prime Therapeutics, Metanomics, Chugai Pharmaceuticals Co., Ltd. and Novartis; he has also filed a patent for the detection of antibodies and T cells against KIR4.1 in a subpopulation of patients with MS and genetic determinants of neutralizing antibodies to interferon-beta; K Rammohan has received honoraria for participating in advisory boards and consulting for Acorda, Biogen, EMD Serono, Genentech, Inc./F. Hoffmann-La Roche Ltd., Genzyme, and Teva; he has also received grants from Accera; G Giovannoni has received honoraria from AbbVie, Bayer HealthCare Pharmaceuticals, Biogen, Canbex Therapeutics, Five Prime Therapeutics, Genzyme, GSK, GW Pharma, Merck, Merck Serono, Novartis, Protein Discovery Laboratories, F. Hoffmann-La Roche Ltd., Synthon, Teva Neuroscience, UCB and Vertex; research grant support from Biogen, Ironwood, Merck Serono, Merz and Novartis; and compensation from Elsevier; J de Seze has received consultancy fees and served as an expert for advisory boards for Alexion, Allergan, Almiral, Bayer, Biogen, Chugai, CSL Behring, Genzyme, LFB, Merck, Novartis, Roche, and Teva; A Bar-Or has received personal compensation for consulting, serving on scientific advisory boards and/or speaking activities from: Bayer, Bayhill Therapeutics, Berlex, Biogen, BioMS, DioGenix, Eli Lilly, Genentech, Inc., GSK, Guthy-Jackson/GGF, Merck Serono, Novartis, Ono Pharmacia, F. Hoffmann-La Roche Ltd., Sanofi-Aventis, Teva Neuroscience and Wyeth.; DL Arnold reports equity interest in NeuroRx Research, which performed the MRI analysis for the trial, and consultation fees from Acorda Therapeutics, Biogen, Genzyme, F. Hoffmann-La Roche Ltd., Innate Immunotherapeutics, MedImmune, Mitsubishi Pharma, Novartis, Receptos, Sanofi Aventis, and Teva; A Sauter is an employee and shareholder of F. Hoffmann-La Roche Ltd.; D Masterman is an employee and/or shareholder of Genentech, Inc. Group, a member of the Roche Group; P Fontoura is an employee and shareholder of F. Hoffmann-La Roche Ltd.; P Chin is an employee and/or shareholder of Genentech, Inc., a member of the Roche Group; Hideki Garren is an employee and shareholder of F. Hoffmann-La Roche Ltd.; J Wolinsky has received compensation for service on steering committees or data monitoring boards for Novartis, F. Hoffmann-La Roche Ltd., Genzyme, and Teva Pharmaceuticals; consultant fees from AbbVie, Actelion, Alkermes, Athersys, Inc., EMD Serono, Forward Pharma, Genentech, Inc., Genzyme (Sanofi), Novartis, F. Hoffmann-La Roche Ltd., Teva, and XenoPort; research support from Genzyme, Sanofi, the NIH, and the NMSS through the University of Texas Health Science Center at Houston (UTHSCH); and royalties for monoclonal antibodies out-licensed to Chemicon International through UTHSCH.

ACKNOWLEDGMENTS

We would like to thank all patients, their families, and the investigators who participated in this trial. This research was funded by F. Hoffmann-La Roche Ltd., Basel, Switzerland. Support for third-party writing assistance for this presentation was provided by F. Hoffmann-La Roche Ltd., Basel, Switzerland.

REFERENCES

1. Miller DH, Leary SM. *Lancet Neurol* 2007;6:903–12
2. Wingerchuk DM, Carter JL. *Mayo Clin Proc* 2014;89:225–40
3. Comi G. *Mult Scler J* 2013;19:1428–36
4. Hartung HP, *et al. Expert Rev Neurother* 2011;11:351–62
5. Polman CH, *et al. Ann Neurol* 2005;58:840-6
6. Hauser SL, *et al. ECTRIMS*, Barcelona, Spain; 7–10 October 2016; abstract 190

Presented at the Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum 2016; February 18-20, 2016; New Orleans, LA

Poster P023